Pharmacodynamics and pharmacokinetics of SO-C101 in cynomolgus monkeys

Irena Adkins[1,2], Nada Podzimkova[1], Natalia Kalynovska[1], Jan Praslicka[1], Guy de Martynoff[3], David Bechard[3], Radek Spisek[1,2], Ulrich Moebius[1]

[1]SOTIO a.s, Jankovcova 1518/2, Prague 7, 170 00, Czech Republic; [2]Department of Immunology, 2nd Faculty of Medicine and University Hospital Motol, Charles University, V Uvalu 84, Prague 5, 150 06, Czech Republic; [3]Cytune Pharma, Nantes F-44300, France

Introduction

SO-C101 is a superagonist fusion protein of interleukin (IL)-15 and the IL-15 receptor α (IL-15Rα) sushi+ domain. SO-C101 effectively stimulates natural killer (NK) cells and memory CD8⁺ T cells with no significant expansion and activation of regulatory T cells which translates to anti-tumor efficacy in mice. In this study, we investigated different administration schedules of SO-C101 in cynomolgus monkeys to assess its pharmacodynamic and pharmacokinetic properties using intravenous (IV) and subcutaneous (SC) routes of administration. SC administration of SO-C101 was more effective than IV administration in activating target immune cells which was correlated to the differences in SO-C101 exposure. Repeated administration of SO-C101 over two weeks promoted an increase of absolute lymphocyte counts and of the circulating NK and CD8⁺ T cell numbers. Moreover, two administrations on consecutive days were optimal and comparable to four daily administrations. We further determined an optimal schedule for a repetitive SO-C101 SC administration to achieve a cycle-dependent stimulation of NK and CD8⁺ T cells over the course of 10 weeks. These studies allowed to correlate the concentration to response relationship *in vitro* with the relationship between C_{max} following SC administration and the resulting NK and CD8⁺ T cell activation levels *in vivo*. Since the potency of SO-C101 to activate NK and CD8⁺ T cells *in vitro* is equivalent between human and cynomolgus monkeys, these studies informed on the dose and schedule selection for the ongoing Phase I clinical study in patients with advanced solid tumors (NCT04234113).

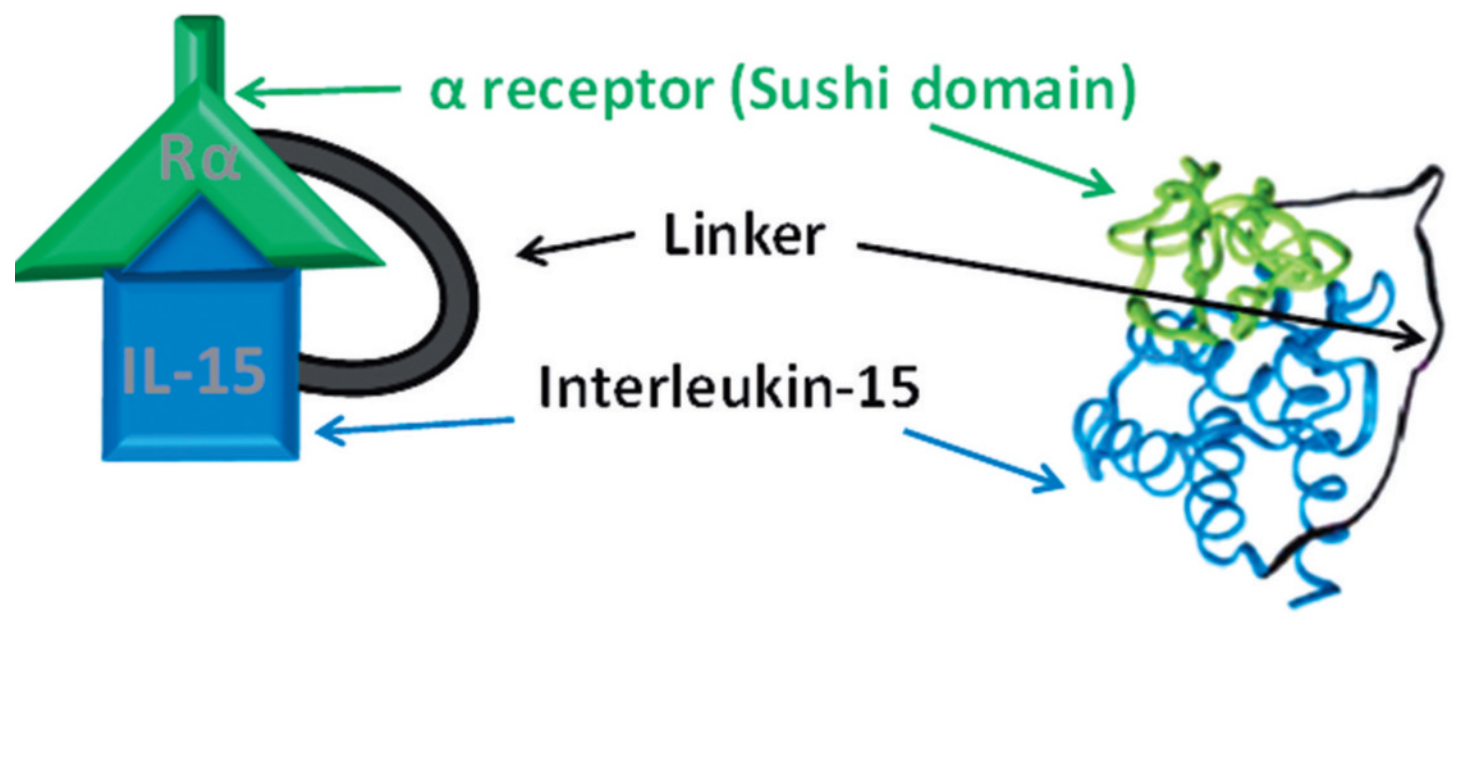

α receptor (Sushi domain)
Rα
Linker
IL-15
Interleukin-15

- SO-C101 (211 amino acids) is a fusion protein that consists of the N-terminal sushi⁺ domain of the human IL-15Rα (77 amino acids) covalently coupled via a non-immunogenic linker of glycine-serine residues (20 amino acids) to the mature IL-15 sequence (114 amino acids) (Mortier et al., 2006).
- SO-C101 acts as a selective and potent agonist of the IL-15 pathway through IL-15Rβγ interaction, thereby inducing proliferation and activation of memory CD8⁺ T cells, NK cells, γδ T cells and NKT cells *in vitro* and *in vivo* and exerts significantly increased anti-tumor efficacy in various mouse cancer models compared to IL-15 (Bessard et al., 2009; Desbois et al., 2016)

Figure 1

SO-C101 induces proliferation of human and cynomolgus monkey NK and CD8⁺ T cells with comparable potency *in vitro*

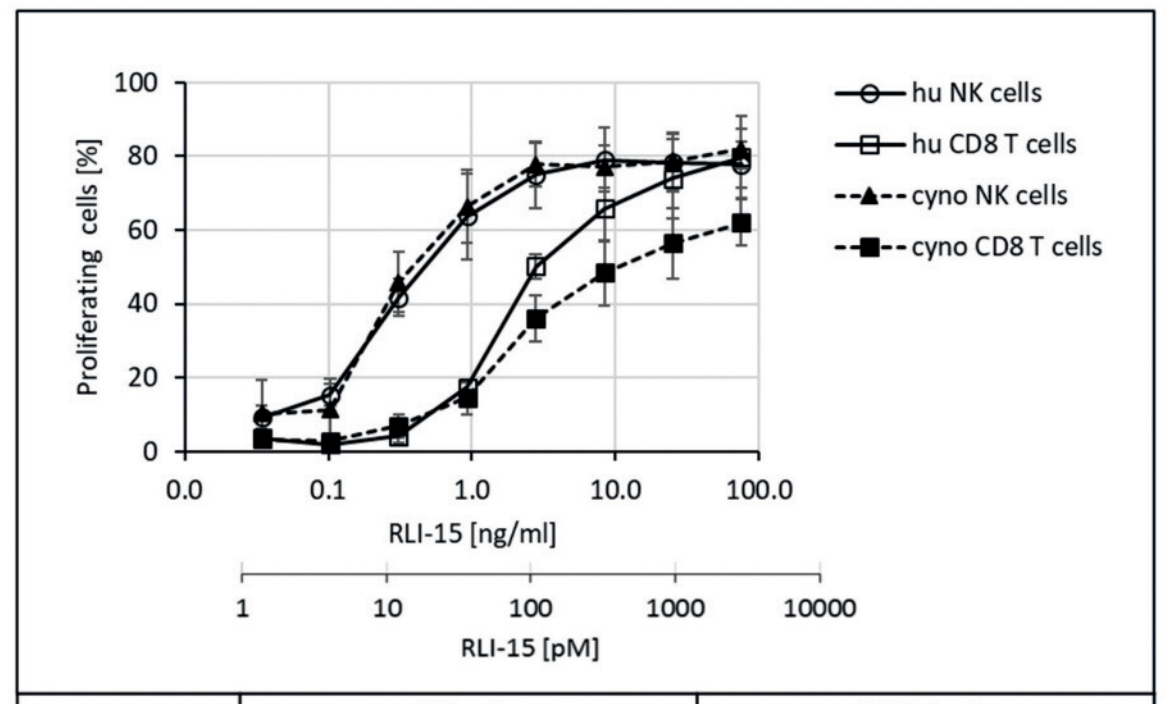

Proliferating cells [%]
RLI-15 [ng/ml]
RLI-15 [pM]
hu NK cells
hu CD8 T cells
cyno NK cells
cyno CD8 T cells

	NK cells		CD8 T cells	
	Human	Cynomolgus	Human	Cynomolgus
EC10 [pM]	2.8	3.7	19.3	14.6
EC50 [pM]	13.9	12.7	86.9	104.0
EC90 [pM]	79.2	46.5	502.9	775.4

Figure 1. SO-C101 stimulates human and cynomolgus monkey PBMC with comparable potency. Peripheral blood mononuclear cells (PBMC) from three different human donors and 3 cynomolgus monkeys were labelled with 5(6)-Carboxyfluorescein diacetate N-succinimidyl ester (CFSE) and subsequently incubated *in vitro* with increasing SO-C101 concentrations for 6 days. Proliferating cells showing a decrease in CFSE fluorescence intensity within the NK cell (NKp46⁺/CD3⁻, human; CD56⁺/CD3⁻, cynomol-gus) and CD8⁺ T cell (CD3⁺/CD8⁺) population were determined by flow cytometry. EC10, EC50, and EC90 values are shown.

Figure 2

SO-C101 administered SC is more effective in activating NK and CD8⁺ T cells than IV administered SO-C101

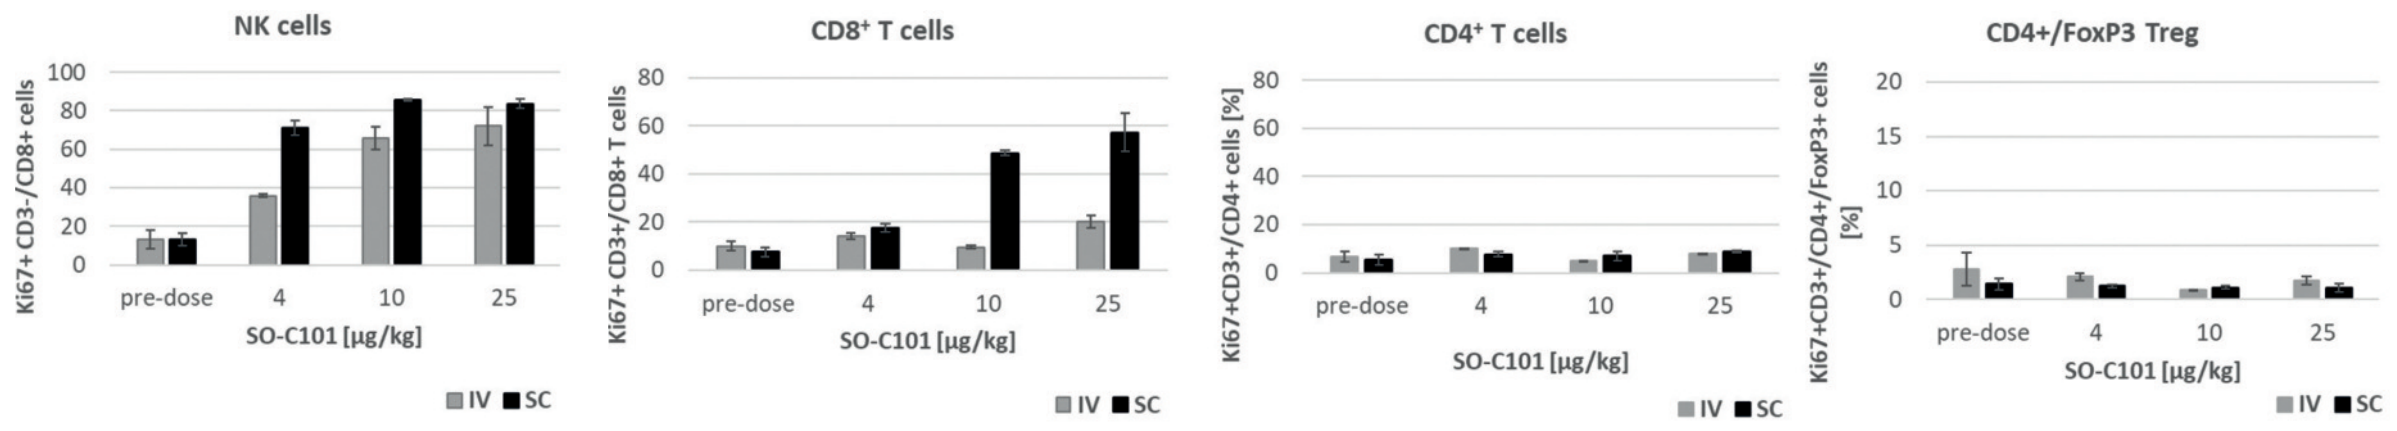

NK cells
CD8⁺ T cells
CD4⁺ T cells
CD4+/FoxP3 Treg
pre-dose
SO-C101 [μg/kg]
IV
SC

Figure 2. Proliferation (Ki67⁺) of NK and CD8⁺ T cells, CD4⁺ T cells and CD4⁺/Foxp3⁺ regulatory T cells after 4 consecutive days of intravenous (IV, 60 min) (grey bars) or subcutaneous (SC) (black bars) treatment with SO-C101 doses as indicated was determined on Day 5 by flow cytometry. NK and CD8⁺ T cell activation by SO-C101 was stronger following SC administration compared to IV administration, resulting in a higher frequency of Ki67⁺CD8⁺ T cell population. Importantly, there was no activation of CD4⁺ T cells and regulatory T cells. This could be attributed to a difference in SO-C101 pharmacokinetics as SC injection of SO-C101 resulted in a longer time period of bioactive SO-C101 concentrations in serum samples compared to IV infusion, see Fig. 3. (n = 2 animals per dose group).

Figure 3

Pharmacokinetics of SO-C101 differs following SC and IV administration

A

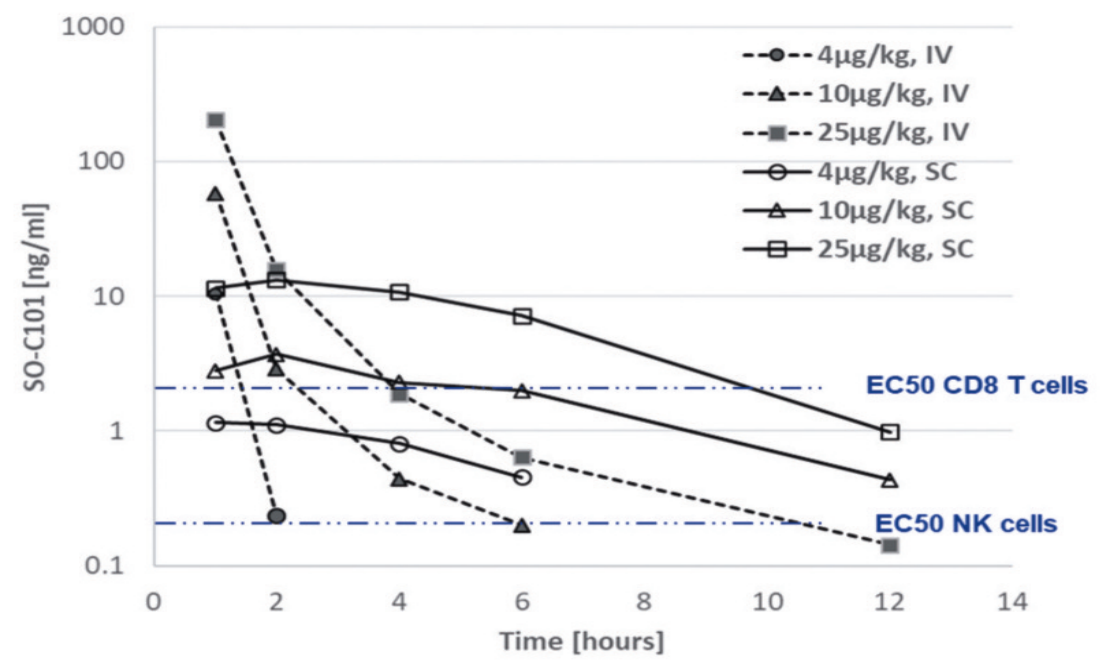

SO-C101 [ng/ml]
Time [hours]
4μg/kg, IV
10μg/kg, IV
25μg/kg, IV
4μg/kg, SC
10μg/kg, SC
25μg/kg, SC
EC50 CD8 T cells
EC50 NK cells

B

Route	Dose μg/kg	C_{max} ng/ml	C_{max} / dose	t_{max} h	$AUC_{(0\text{-}12h)}$ h*ng/ml	$AUC_{(0\text{-}12h)}$ / dose
IV	4	10.47	2.62	1.0	10.82	2.70
IV	10	58.40	5.84	1.0	64.40	6.44
IV	25	204.5	8.18	1.0	234.6	9.38
SC	4	1.19	0.30	1.50	5.12	1.28
SC	10	3.73	0.37	2.00	22.27	2.23
SC	25	14.35	0.57	2.50	84.57	3.38

Figure 3. (A) Pharmacokinetic assessment following a single IV infusion (60 min) or a single SC injection of SO-C101 at 4 μg/kg, 10 μg/kg and 25 μg/kg, respectively (n = 2). EC50 for NK and CD8⁺ T cells in blue. **(B)** SO-C101 C_{max} values were reached following the 1-hour IV infusion consistently at the end of infusion, whereas for SC injection C_{max} was reached between 1 h and 4 h after injection. C_{max} and $AUC_{(0\text{-}12h)}$ increased with dose in both, the IV and SC dosing groups. This increase was more than dose-proportional for both, C_{max} and $AUC_{(0\text{-}12h)}$ and was more pronounced following IV than SC administration. The absolute bioavailability of SO-C101 after SC injection ranged between about 35% at 10 μg/kg and 25 μg/kg and about 47% at 4 μg/kg. The half-life of SO-C101 was 3-4 h consistent over the range of all doses tested.

Figure 4

SO-C101 enhances expression of immune cell specific genes in the skin of cynomolgus monkeys upon SC treatment

A

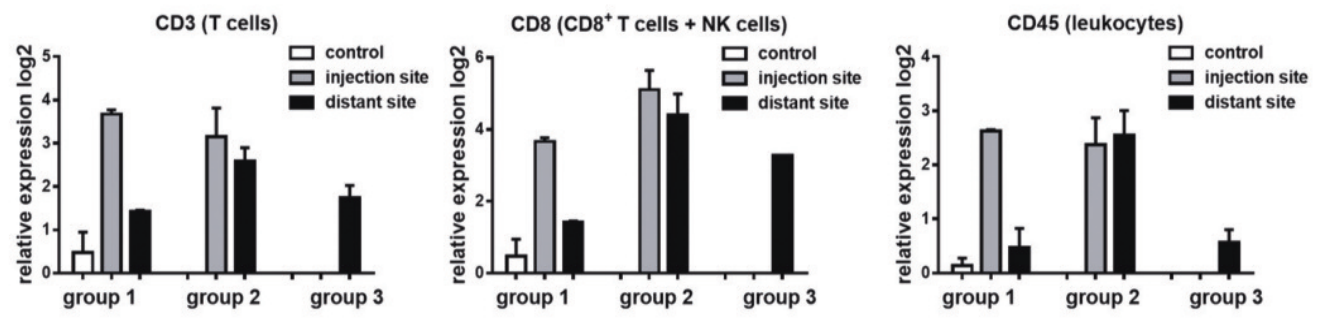

CD3 (T cells)
CD8 (CD8⁺ T cells + NK cells)
CD45 (leukocytes)
control
injection site
distant site
relative expression log2
group 1
group 2
group 3

B

Group	Route	Dosing days	Dose/adm (μg/kg)	Route	Dosing days	Dose/adm (μg/kg)
1	-	Not treated		SC	22, 23, 29, 30, 36, 37	15
2	SC	1, 2, 3, 4	25	SC	22 – 25, 29 – 32	15
3	IV	1, 2, 3, 4	25	IV	22 – 25, 29 – 32	40

Figure 4. (A) qPCR analysis of CD45, CD3 and CD8a immune cell genes from skin biopsies of cynomolgus monkeys collected at the injection site and a distant site at Day 40. Biopsies were collected 8 days (Group 2 and 3) and 2 days after the last treatment (Group 1), respectively. Control biopsies were obtained from untreated monkeys. Group 3 showed the lowest skin immune cell infiltration which can be most likely attributed to a lower level of SO-C101 stimulation induced by IV administration. In group 1, the immune cell infiltration was significantly higher in samples from the injection sites than in samples from distant sites in contrast to group 2 where the expression of CD3 and CD8a genes was comparable between injection and distant site samples. This might be correlated with the denser SO-C101 administration schedule. **(B)** The biopsy groups' treatment schedule (n = 2).

Figure 5

Two SC administration of SO-C101 for two consecutive weeks lead to an optimal NK and CD8⁺ T cell activation

A

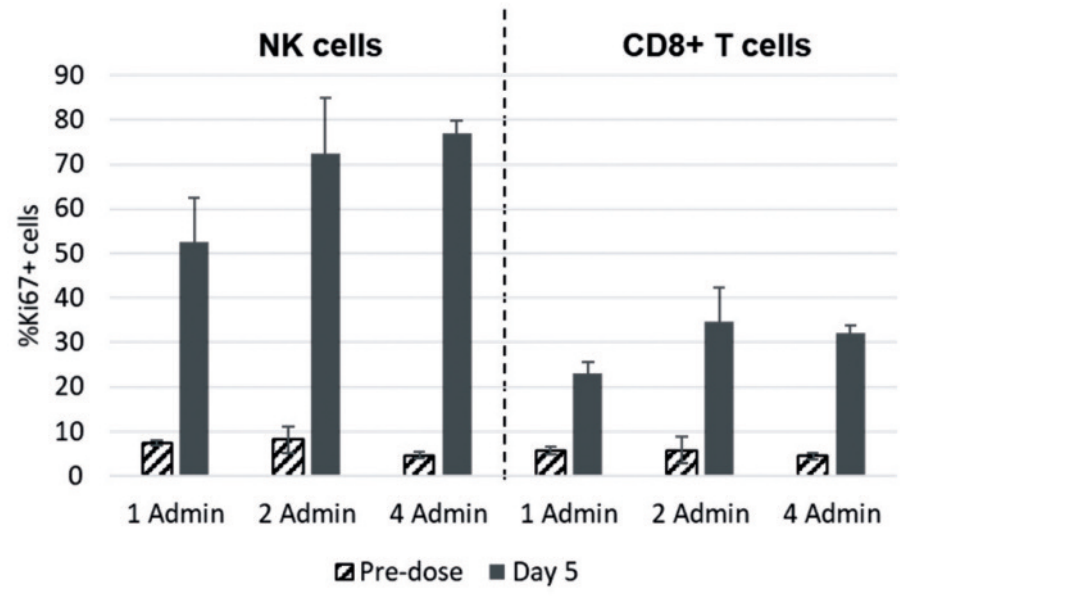

NK cells
CD8+ T cells
%Ki67+ cells
1 Admin
2 Admin
4 Admin
Pre-dose
Day 5

B

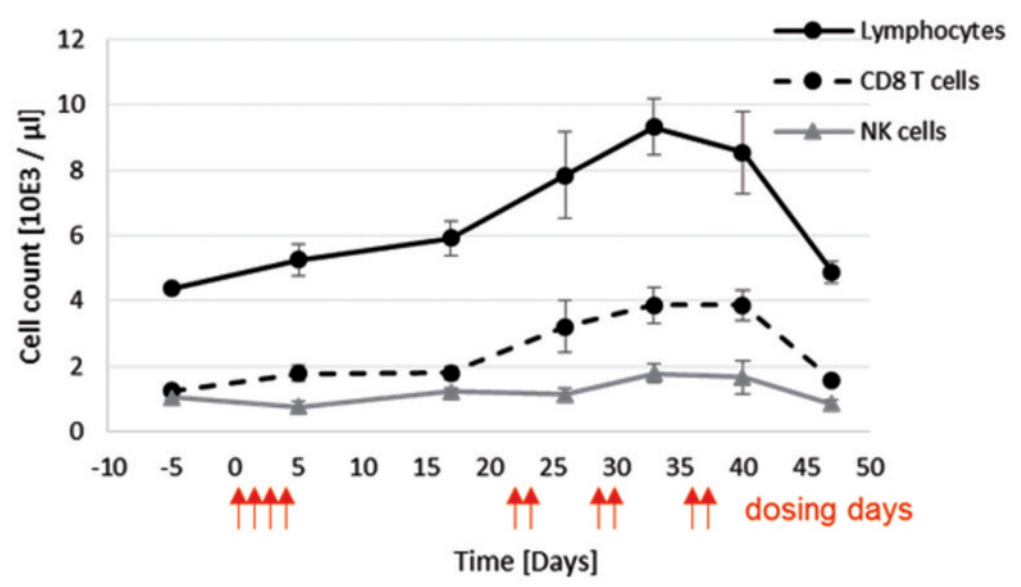

Lymphocytes
CD8 T cells
NK cells
Cell count [10E3 / μl]
Time [Days]
dosing days

Figure 5. (A) NK and CD8⁺ T cells after SC injection of 15 μg/kg SO-C101 were analyzed on Day 5 by flow cytometry. 1 Admin: single SC administration on Day 1; 2 Admin: two SC administrations on Days 1 and 2; 4 Amin: four SC administrations on Days 1-4. The fraction of Ki67⁺ cells was determined in the CD3⁻/CD8⁺/CD45⁺ (NK cells) and CD3⁺/CD8⁺/CD45⁺ (CD8⁺ T cells) cell fraction. **(B)** Cynomolgus monkeys were dosed with daily SC administrations (Day 1 – 4) at 10 μg/kg. Following a treatment-free period of two weeks, animals were dosed on two consecutive days over three weeks (study Days 22, 23, 29, 30, 36 and 37, shown by arrows). Lymphocyte analysis was performed on day 5 of each dosing week (Days 5, 26, 39 and 46). Lymphocyte counts were determined during hematology assessment. NK and CD8⁺ T cells were determined by flow cytometry and multiplication of their relative proportion within CD45⁺ cells with total white blood cell counts. The data show cell count increase within first two weeks of treatment after the day 20. There is no benefit of the third dosing week on immune cell counts in the blood. (n = 2-4 animals per dose group).

Figure 6

Pulsed cycle regimen of SO-C101 administration with treatment gaps leads to an optimal NK and CD8⁺ T cell stimulation

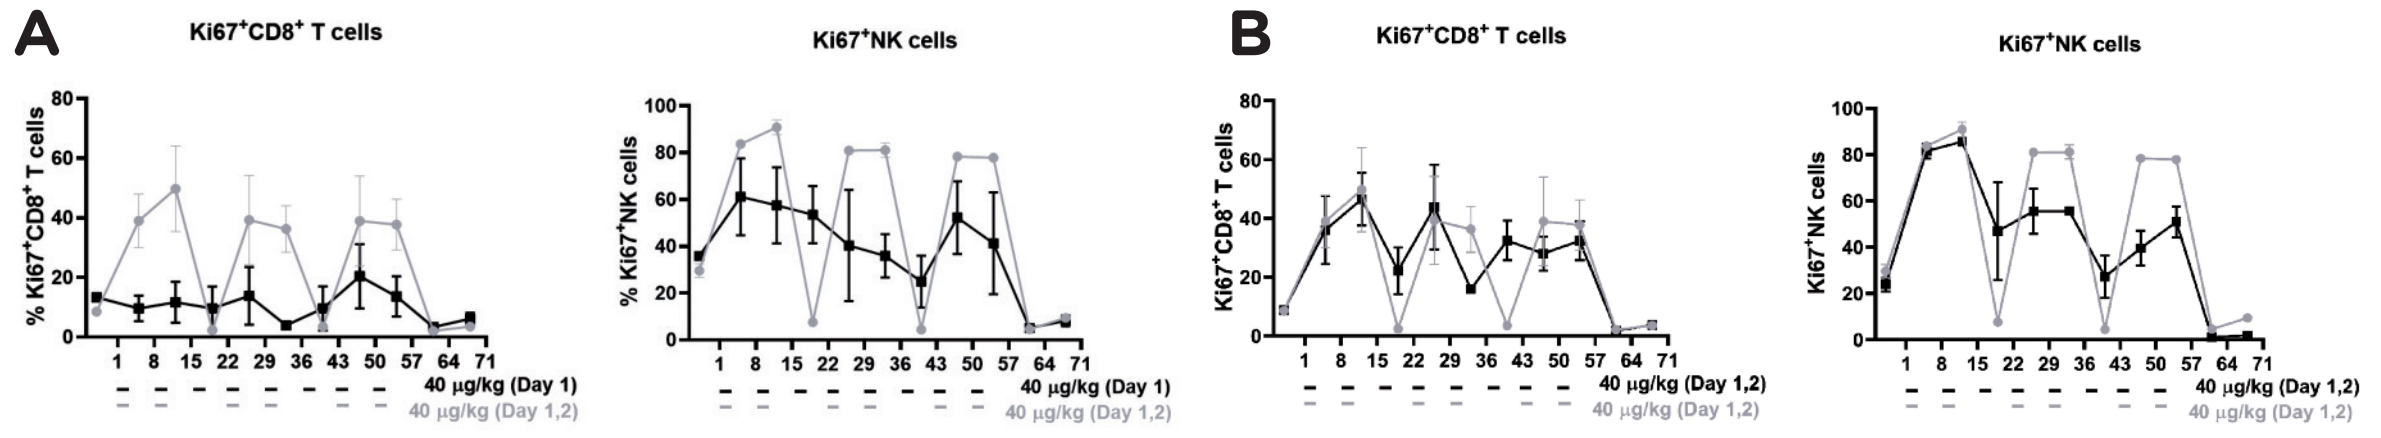

A
Ki67⁺CD8⁺ T cells
Ki67⁺NK cells
% Ki67⁺CD8⁺ T cells
% Ki67⁺NK cells
40 μg/kg (Day 1)
40 μg/kg (Day 1,2)
B
Ki67⁺CD8⁺ T cells
Ki67⁺NK cells
40 μg/kg (Day 1,2)
40 μg/kg (Day 1,2)

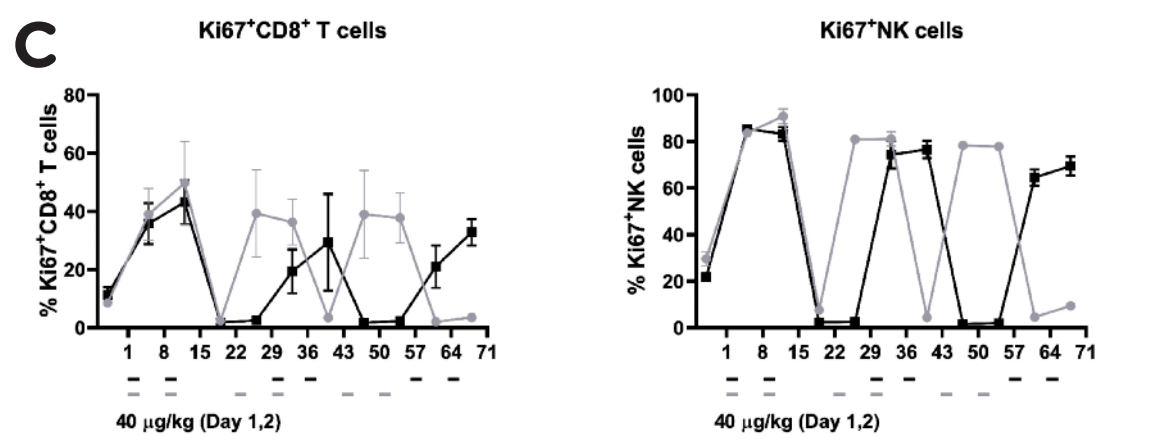

C
Ki67⁺CD8⁺ T cells
Ki67⁺NK cells
% Ki67⁺CD8⁺ T cells
% Ki67⁺NK cells
40 μg/kg (Day 1,2)

Figure 6. 10-week scheduling study was conducted with 40 μg/kg SO-C101 as an administration dose. SO-C101 was administered once daily in a continuous schedule (every week at the same day 1) or as a gap schedule (1 or 2 weeks treatment pause as depicted by lines under graphs, two same days a week) **(A)** Continuous weekly (Day 1) treatment leads to a decrease of NK and CD8⁺ T cell proliferation in contrast to gap (1 week) (Day 1,2) treatment **(B)** Continuous weekly (Day 1,2) treatment leads to a decrease of mainly NK proliferation in contrast to gap (1 week) (Day 1,2) treatment. **(C)** Gap treatment schedule 1 or 2 weeks (Day 1,2 leads to an optimal NK and CD8⁺ T cell proliferation. The data favor pulsed cycle regimen of SO-C101 administration with treatment gaps 1 or 2 weeks. Data represent mean + SEM from 2 monkeys per group.

Conclusions

- **SO-C101 activates human and cynomolgus NK and CD8⁺ T cell proliferation with a similar potency *in vitro***
- **Subcutaneous administration of SO-C101 is more effective in activating NK and CD8⁺ T cells in blood and in the skin than intravenous administration. This is related to the difference of SO-C101 pharmacokinetics between the SC and IV routes of administration**
- **Two subcutaneous administrations of SO-C101 for two consecutive weeks lead to an optimal NK and CD8⁺ T cell stimulation**
- **A pulsed cyclic treatment schedule with a resting period of 1 or 2 weeks favors optimal NK and CD8⁺ T cell stimulation compared to continuous treatment without a resting period.**

For more information please contact Irena Adkins, adkins@sotio.com